Establishing a robust exosome manufacturing process for translational studies

Smriti M. Krishna[1], Xenia Sango[1], Louise White[1,2], Craig Newton[1], Johannes F. Weber[1], Ella L. Johnston[1], Mitch Shambrook[1], Ian Nisbet[1], David N. Haylock[1]

[1]VivaZome Therapeutics Pty Ltd, Victoria, Australia [2]SeerPharma Pty Ltd, Victoria, Australia

Introduction

Despite exponential growth and interest in exosome or extracellular vesicle (EV)-based therapeutics, there is no standardised commercial-scale method for exosome-manufacture globally, and no registered EV therapeutics. VivaZome Therapeutics (VZT) via a CRC-P project with partners Cytiva, University of Queensland (UQ), Australia National University (ANU), La Trobe University and SeerPharma is building expertise within Australia to manufacture therapeutic EVs. In this project, VivaZome will establish (1), a purpose-built manufacturing facility for EV manufacture, ready for technology transfer to a GMP manufacturing facility for the production of EV clinical batches for non-clinical toxicology and preclinical studies and (2), a dedicated EV-analytics laboratory (PC2) to provide stage-appropriate analytics for in-process control and product quality assurance.

Translating EVs into Clinical Products

VivaZome Key Elements

Products that have profound beneficial effects for patients and their doctors

A commitment to taking Australian innovation to the world

Utilising the potential of customised EVs for therapeutic purposes

An emphasis on building a valuable IP portfolio and being a scientific leader in our field

Purpose-built Manufacturing Facility

Quality management implementation from SeerPharma will ensure that both the manufacturing cleanroom and analytics laboratory operate according to recognised standards for laboratories (ISO-17025), FDA and OECD standards for GLP and current best Quality Management practices for the development of biotechnological products.

EV Analytics Laboratory

MISEV (Minimal Information for Studies of Extracellular Vesicles) 2018 (2022-2023 revision passes)

EV Production
-Cell source/ Tissue source / Passage
-Cell culture medium
-EV enrichment process

EV Function
-Activity of native EVs (non-cellular)
-Attributes of Cargos (miRNA, DNA, Lipids)

EV Characterization
-Imaging, single EV level
-Size & Quantification (NTA, Flow cytometry)
-Concentration of Proteins & Lipids
-EV associated marker presence

Rigor & standardization

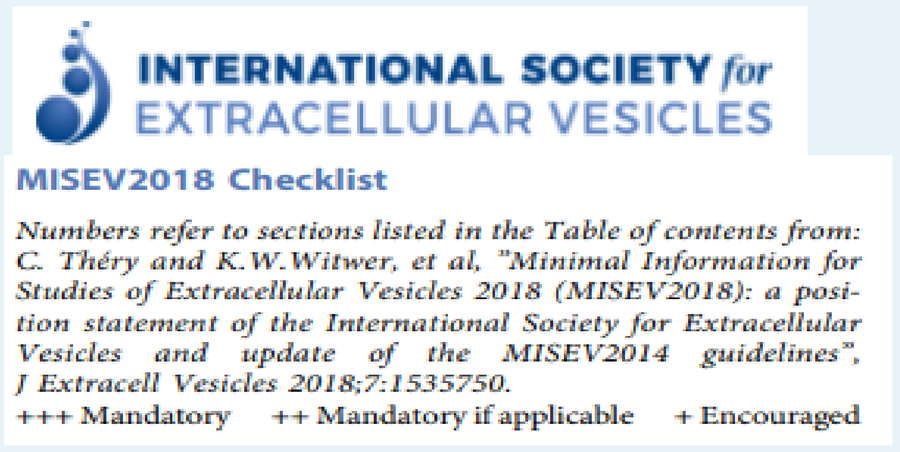
INTERNATIONAL SOCIETY for EXTRACELLULAR VESICLES
MISEV2018 Checklist
Numbers refer to sections listed in the Table of contents from: C. Théry and K.W.Witwer, et al, "Minimal Information for Studies of Extracellular Vesicles 2018 (MISEV2018): a position statement of the International Society for Extracellular Vesicles and update of the MISEV2014 guidelines", J Extracell Vesicles 2018;7:1535750.
+++ Mandatory ++ Mandatory if applicable + Encouraged

Robust Preclinical Studies

- Considerations for choosing the right model for safety and performance testing, use and considerations for "clinically relevant" disease models.

- Strategically plan and leverage preclinical studies to build efficiency into regulatory submissions to save time and money throughout the development continuum.

- Translational projects will adhere to the ARRIVE guidelines (animal research: reporting on *in vivo* experiments) and standards set by the *International Society for Extracellular Vesicles (ISEV)* for collection, processing, testing, quality control, and manufacture of exosomes.

Translational Research

The translational aspect of this project will focus on development and testing of novel customised-EVs with enhanced anti-inflammatory and targeting function, tailored to treat neurological conditions; using robust preclinical-models of traumatic brain injury (TBI) and Age-Related Macular Degeneration (AMD).

AMD

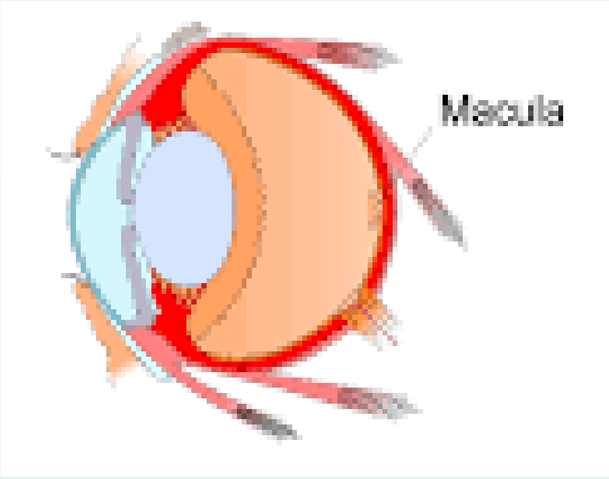
Macula

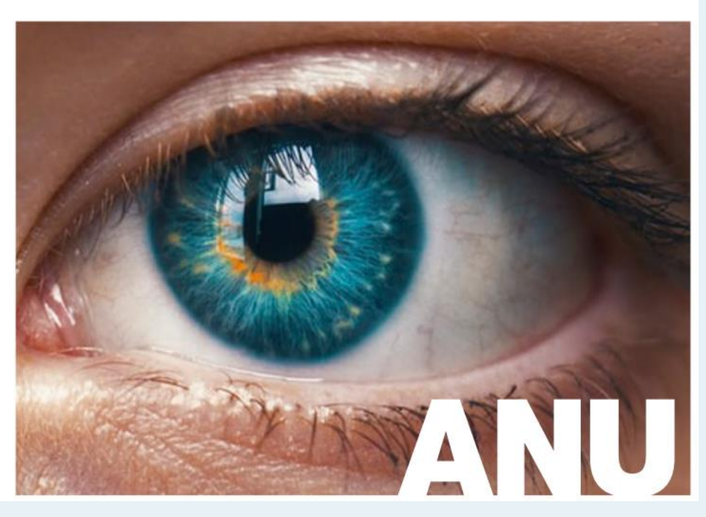
ANU

TBI

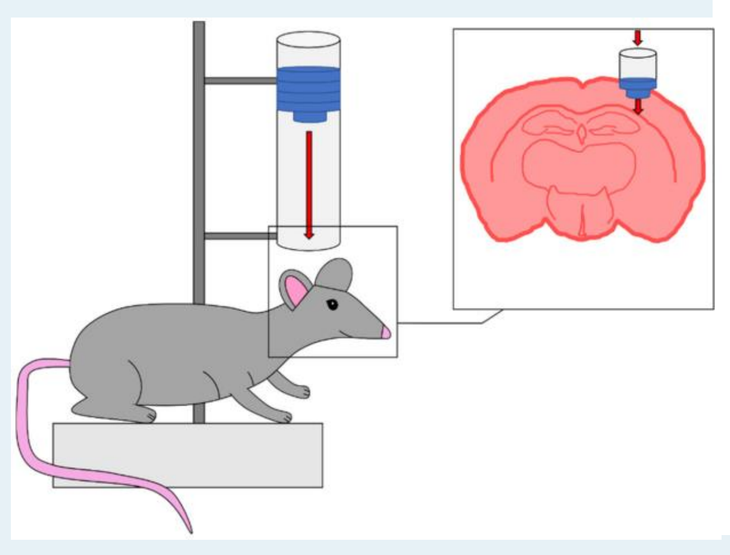

Regulatory Path - EVs to Clinical Trial

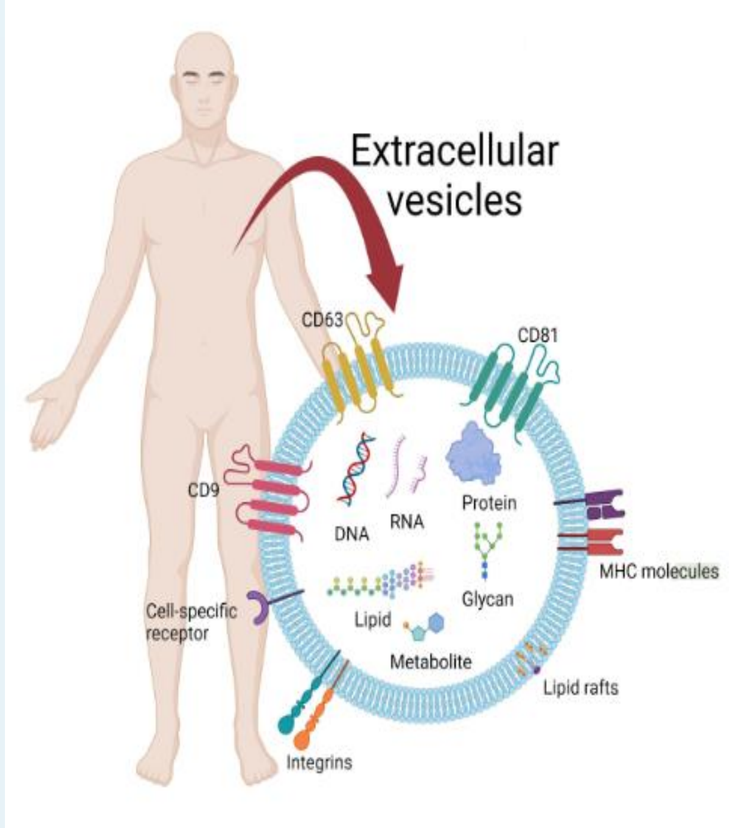
Extracellular vesicles
CD63
CD81
CD9
DNA
RNA
Protein
MHC molecules
Cell-specific receptor
Lipid
Glycan
Metabolite
Lipid rafts
Integrins

Key Dossier: Chemistry Manufacturing & Controls (CMC)

- CMC describes identity, quality & strength of the investigational product

Information includes:

- Source and raw materials and their testing
- Control procedures to assure product quality and robustness
- Appropriate test methods
- Consistent & long-term stability of quality
- Identification & control of critical steps & variables in the manufacturing process

Overview of Exosome Manufacturing for Translation / Development Roadmap

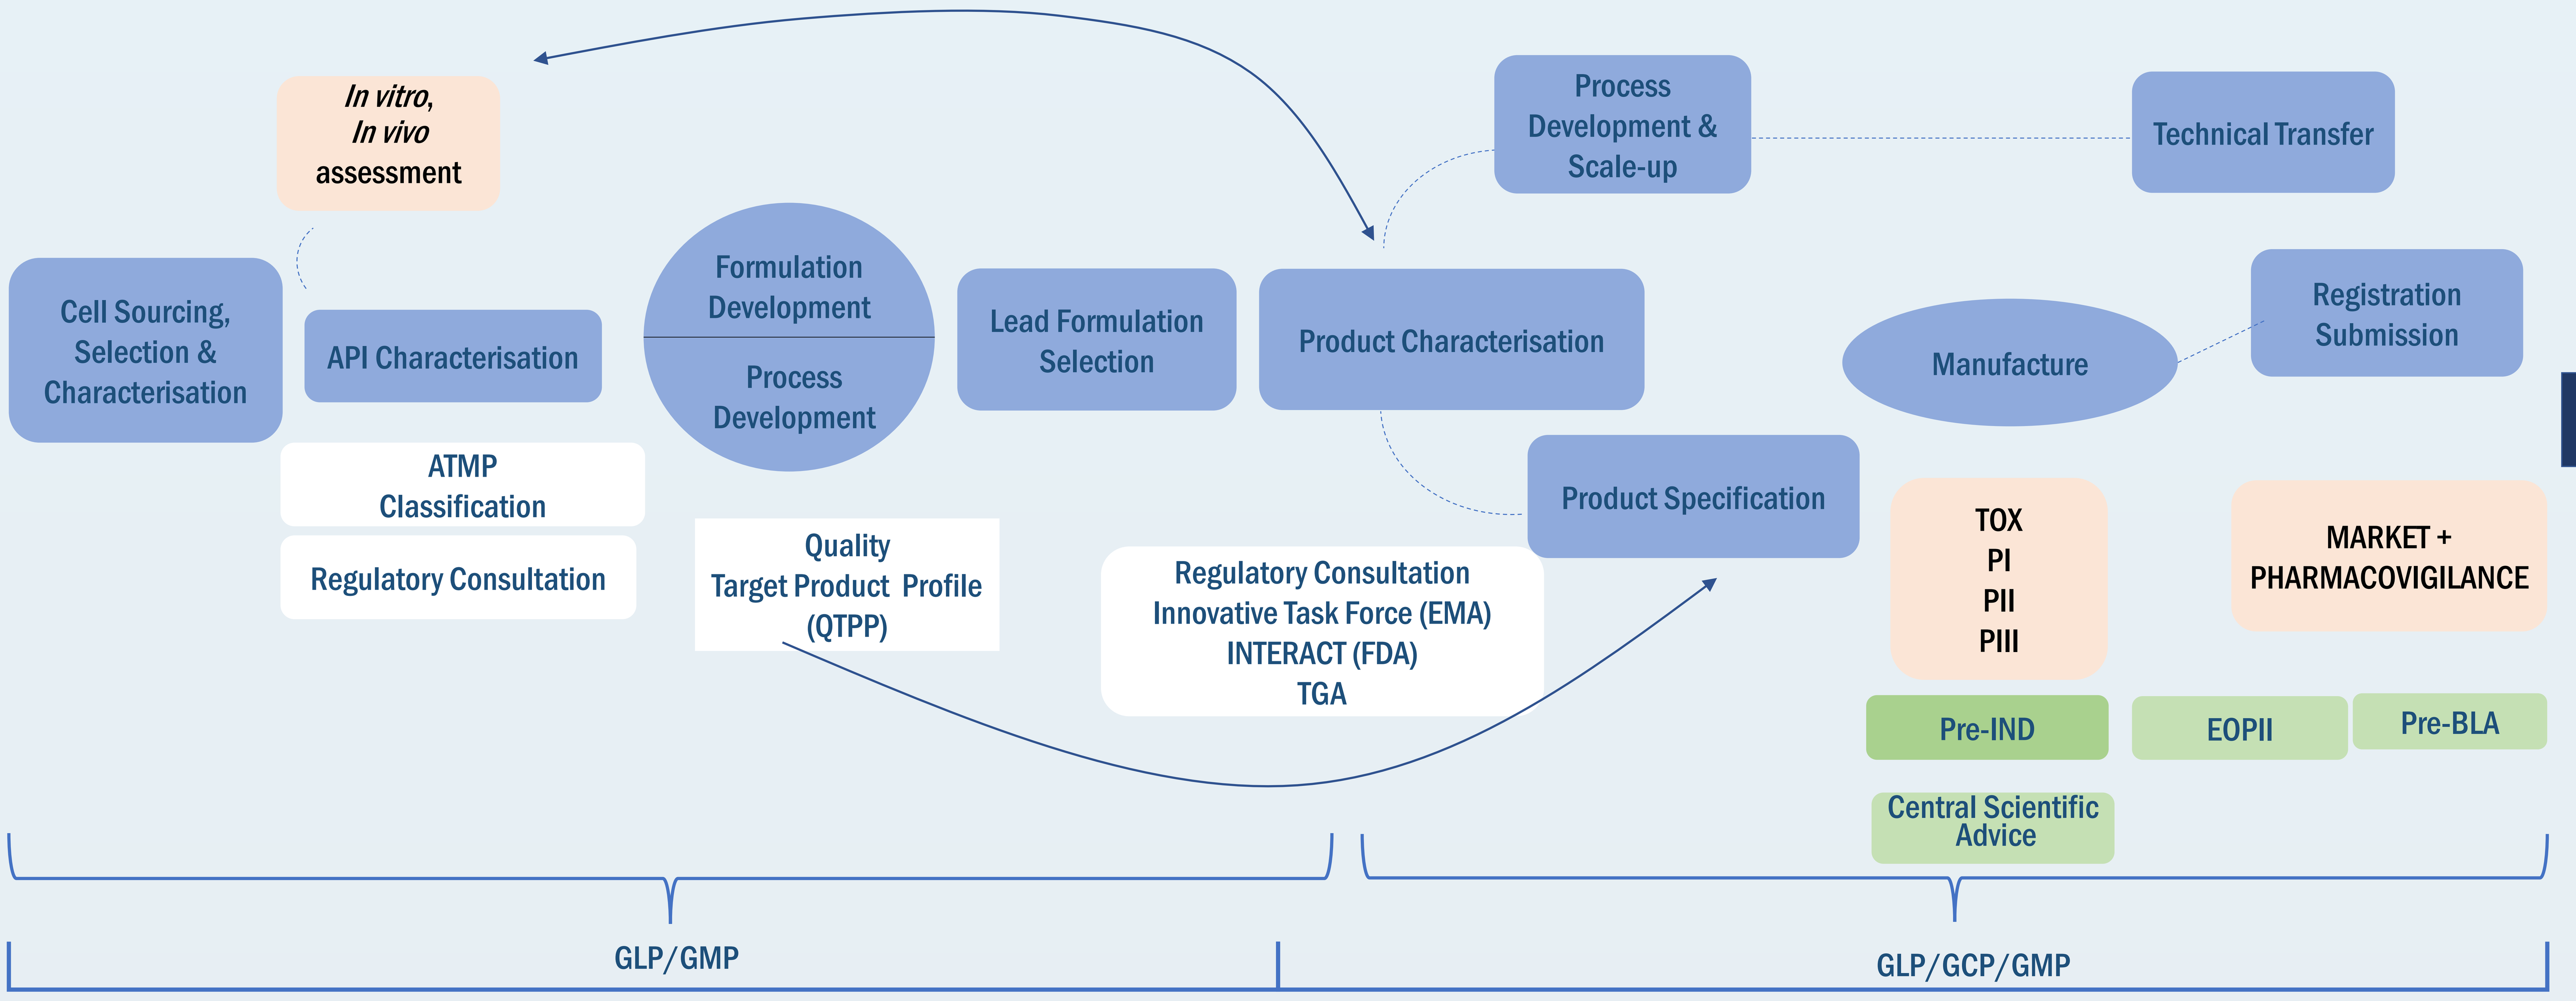
In vitro, In vivo assessment
Cell Sourcing, Selection & Characterisation
API Characterisation
ATMP Classification
Regulatory Consultation
Formulation Development
Process Development
Quality Target Product Profile (QTPP)
Lead Formulation Selection
Product Characterisation
Process Development & Scale-up
Technical Transfer
Product Specification
Regulatory Consultation
Innovative Task Force (EMA)
INTERACT (FDA)
TGA
Manufacture
Registration Submission
TOX
PI
PII
PIII
MARKET + PHARMACOVIGILANCE
Pre-IND
EOPII
Pre-BLA
Central Scientific Advice
GLP/GMP
GLP/GCP/GMP
Quality management systems

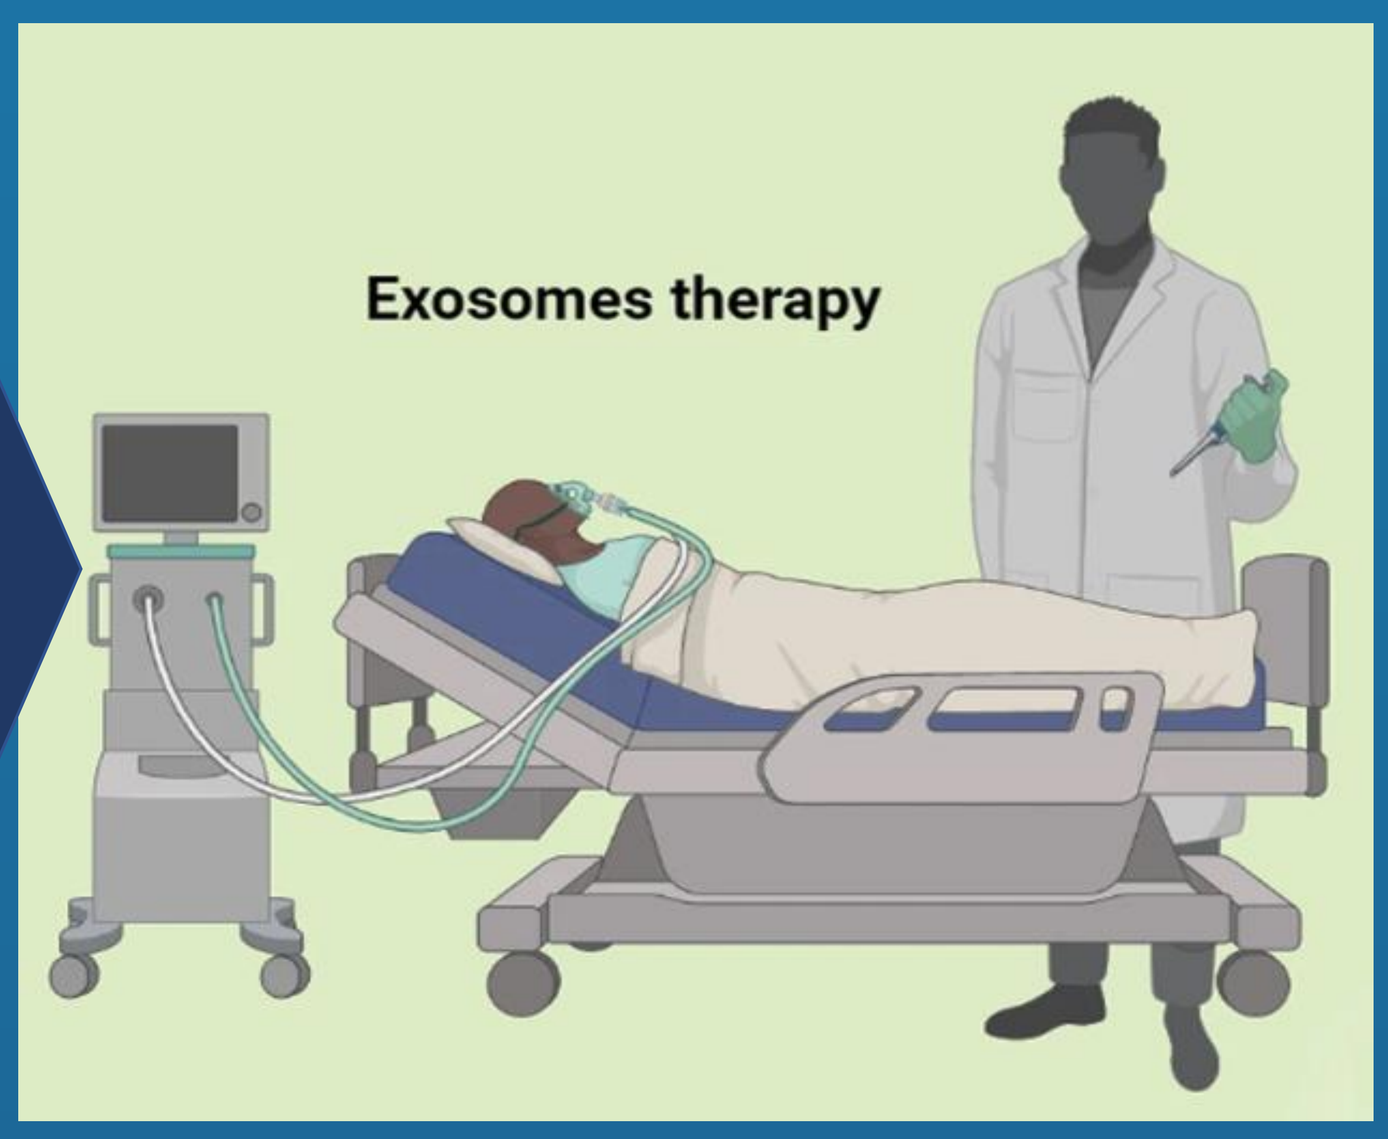
Exosomes therapy

Conclusion

- EVs are a promising platform for the treatment of many diseases. For EVs to be considered a safe, reliable and effective therapeutic, accurate, rapid, cheap, standardized, specific, & easier methods for their separation and purification have to be developed.
- Emphasis on their safety, feasibility, pharmacokinetic and pharmacodynamic characteristics maintained throughout the scalable EV manufacturing process.
- An understanding the **mechanism of action (MoA)** is essential for the clinical translation of therapeutics based on EVs.
- VivaZomes goal is to close the gap of preclinical research and increase product manufacture reproducibility by establishing and using every available scientific, clinical and industry standards to meet regulatory requirements in product manufacture and testing.

© VivaZome Therapeutics Pty Ltd

VivaZome.com